The Healing Power of Nature

Application of Kerecis Graft for Deep Diabetic Foot Ulcer Closure Following Partial Amputation

Randy Semma, DPM, Michele Bertelle-Semma, DPM

Corewell Health Farmington Hills, MI

INTRODUCTION

Diabetic foot wounds remain a major cause of morbidity, often progressing to infection, chronic ulceration, and lower-extremity amputation. Following partial amputations, deep soft-tissue defects can be particularly challenging to manage due to impaired vascularity, neuropathy, and prolonged inflammation. Advanced biologic grafts have emerged as promising adjuncts to standard wound care, offering structural support and promoting tissue regeneration.

Kerecis Omega3 Wound, an acellular fish-skin matrix rich in natural omega-3 fatty acids, has been shown to enhance healing through its unique scaffold and bioactive properties. This case series explores the effectiveness of a single application of Kerecis, used alongside standard wound care and consistent offloading, in facilitating closure of deep post-amputation diabetic foot wounds. The aim is to evaluate its role within a limb-salvage strategy for high-risk patients. The overarching goal is to demonstrate that biologic grafts such as Kerecis can serve as an effective adjunct to standard care by improving healing outcomes in high-risk diabetic foot wounds. By integrating this advanced wound therapy early in the post-surgical phase, the study aims to reduce complications, enhance limb preservation, and ultimately improve patient quality of life.

The purpose of the research is to contribute to the growing body of evidence supporting the clinical use of fish-skin xenografts in diabetic foot management, and to provide data that could support broader adoption in routine practice for limb salvage.

METHOD

This case series included diabetic patients with deep foot wounds after partial amputation. All patients received standard wound care plus one application of Kerecis Omega3 Wound, a fish-skin acellular dermal matrix. After sharp debridement, the graft was trimmed to size, applied to clean, viable, non-infected wound beds, and covered with a non-adherent dressing, sterile gauze, and an elastic compression wrap.

Consistent offloading—using total contact casting or offloading boots—was implemented for all patients. Weekly clinic visits included clinical and photographic evaluations. Healing was monitored through graft integration, granulation formation, wound-size reduction, and full epithelialization until complete closure.

APPLICATION OF KERECIS GRAFT FOR DEEP DIABETIC FOOT ULCER CLOSURE FOLLOWING PARTIAL AMPUTATION

Patient History: A 50-year-old male with no significant medical history presented with a right 5th toe injury sustained on 11/07/2024. Pain progressively worsened. He visited the ED on 11/12/2024, where X-rays showed no acute findings, and he was discharged. Symptoms continued to worsen, with increasing pain, swelling, and skin breakdown. He returned to the ED on 11/14/2024 due to escalating pain and swelling extending to the dorsal foot.

Wound History: This case series evaluated diabetic patients with deep foot wounds following partial amputations. All patients received standard wound care plus a single application of Kerecis Omega3 Wound, a fish-skin–derived acellular dermal matrix. The graft was applied after sharp debridement to clean, viable, non-infected wound beds.

The Kerecis graft was trimmed to fit and placed directly onto the wound, then covered with a non-adherent dressing, sterile gauze, and secured with an elastic compression wrap. Offloading methods—such as total contact casting or offloading boots—were used consistently to minimize mechanical stress.

Patients were followed weekly with clinical and photographic assessments. Healing outcomes included graft integration, granulation formation, wound size reduction, and full epithelialization, tracked until complete wound closure.

Patient outcomes: Patient went on to completely heal with no further complications.

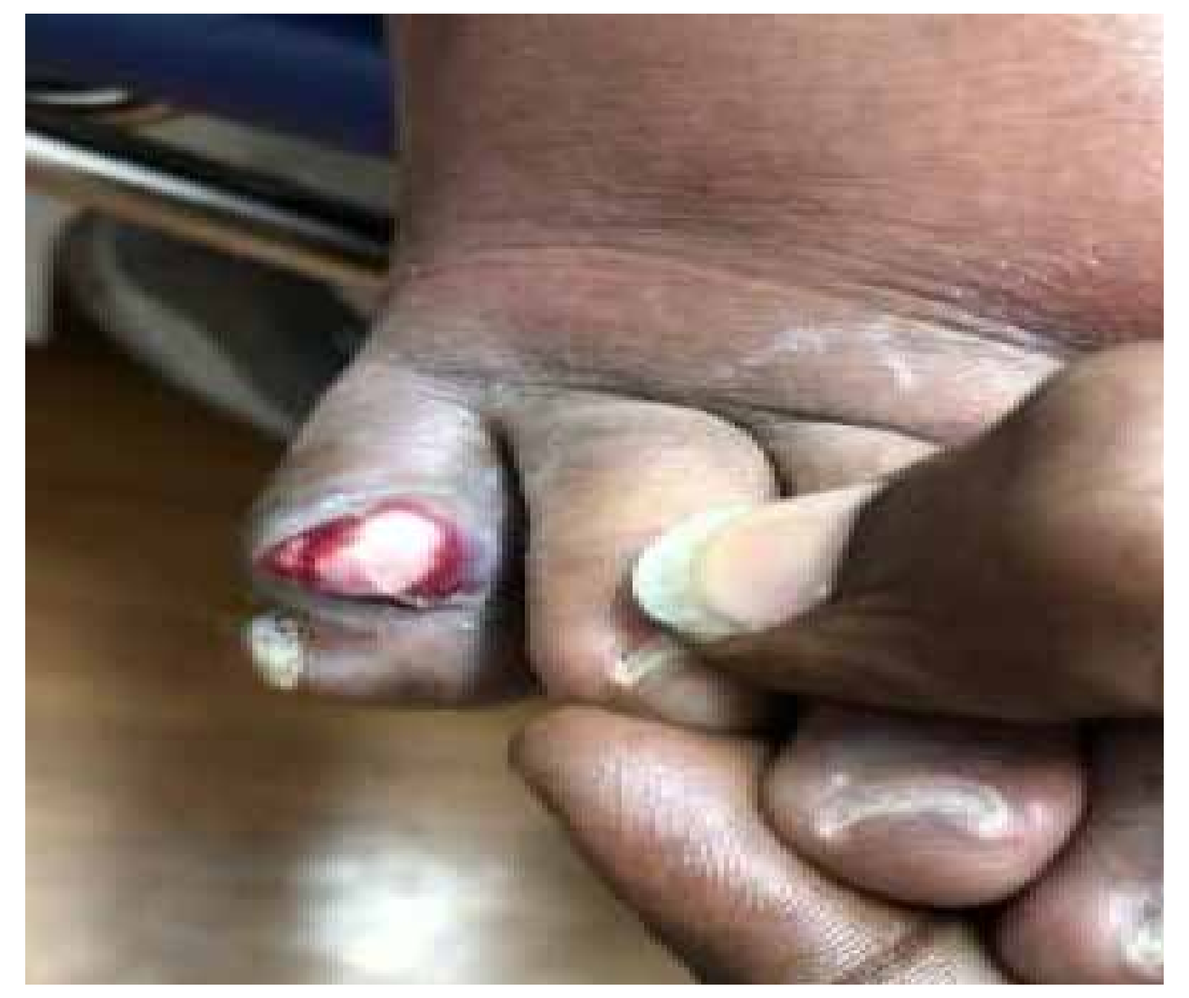

11/12/24 INITIAL ER PRESENTATION

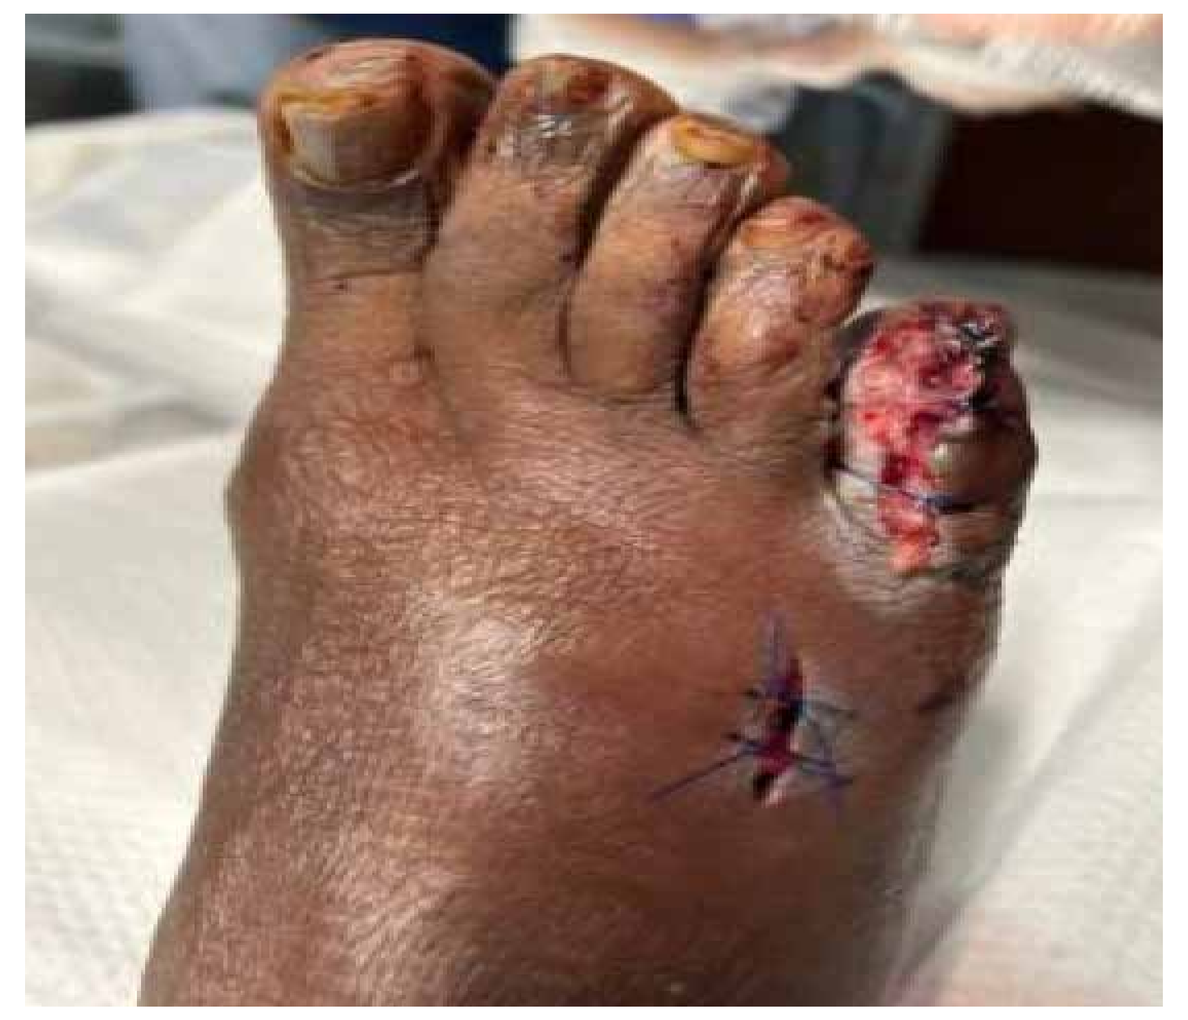

11/15/24 S/P INCISION AND DRAINAGE OF THE FIFTH TOE

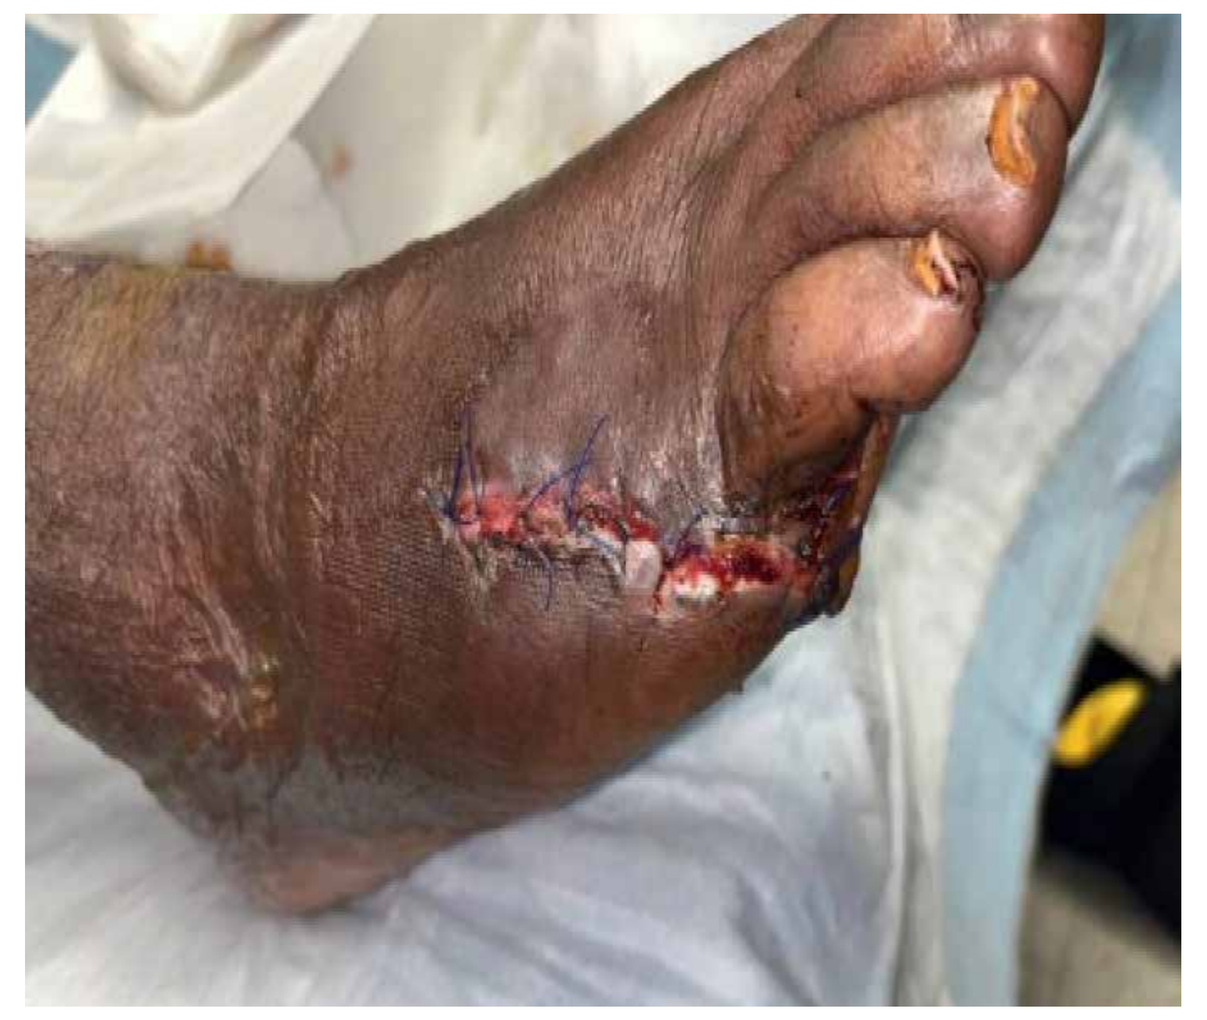

11/17/24 S/P PARTIAL FIFTH RAY AMPUTATION

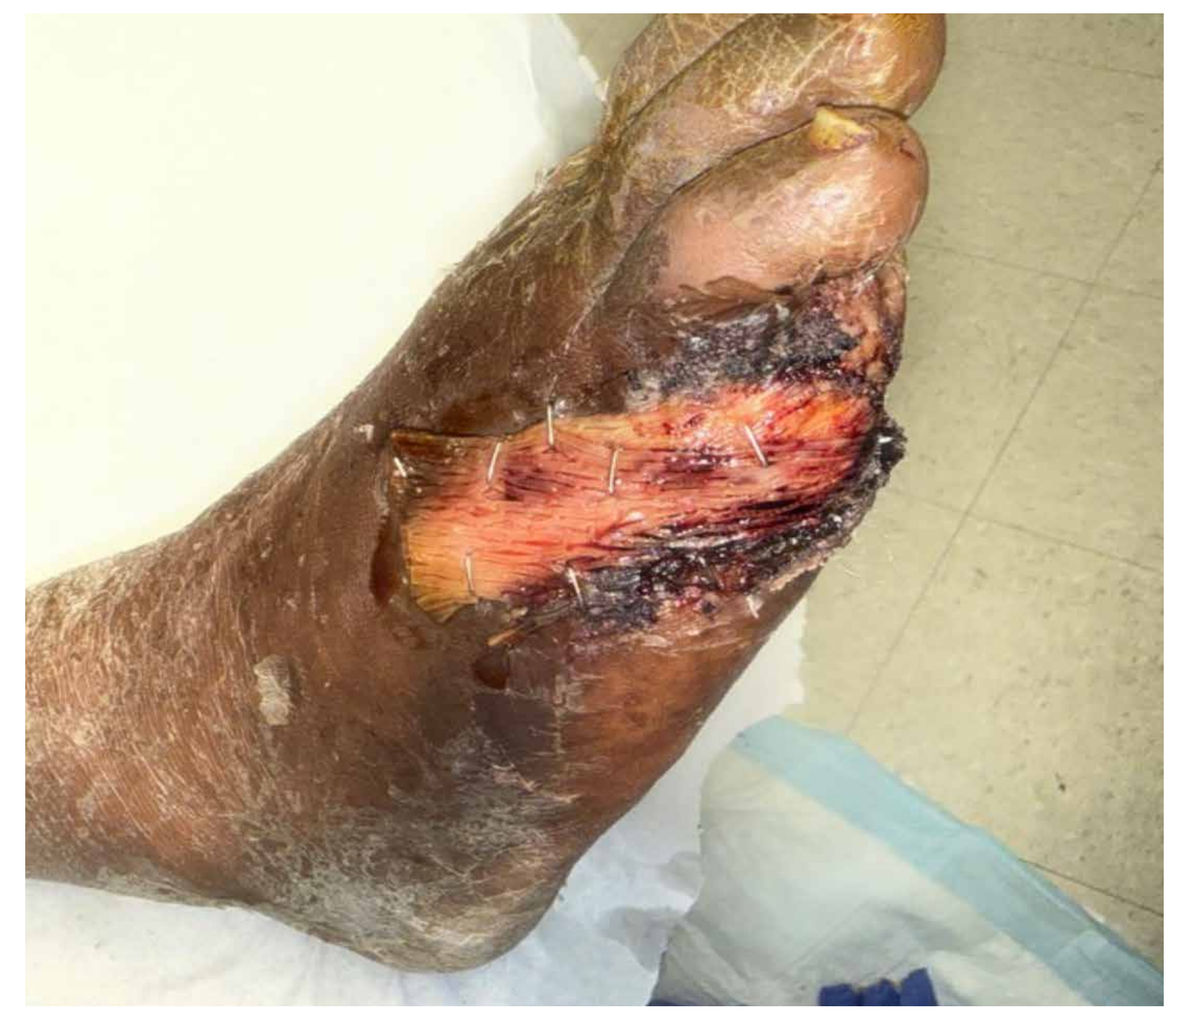

1/8/25 (2 WEEKS S/P DEBRIDEMENT AND APPLICATION OF KERECIS SKIN GRAFT

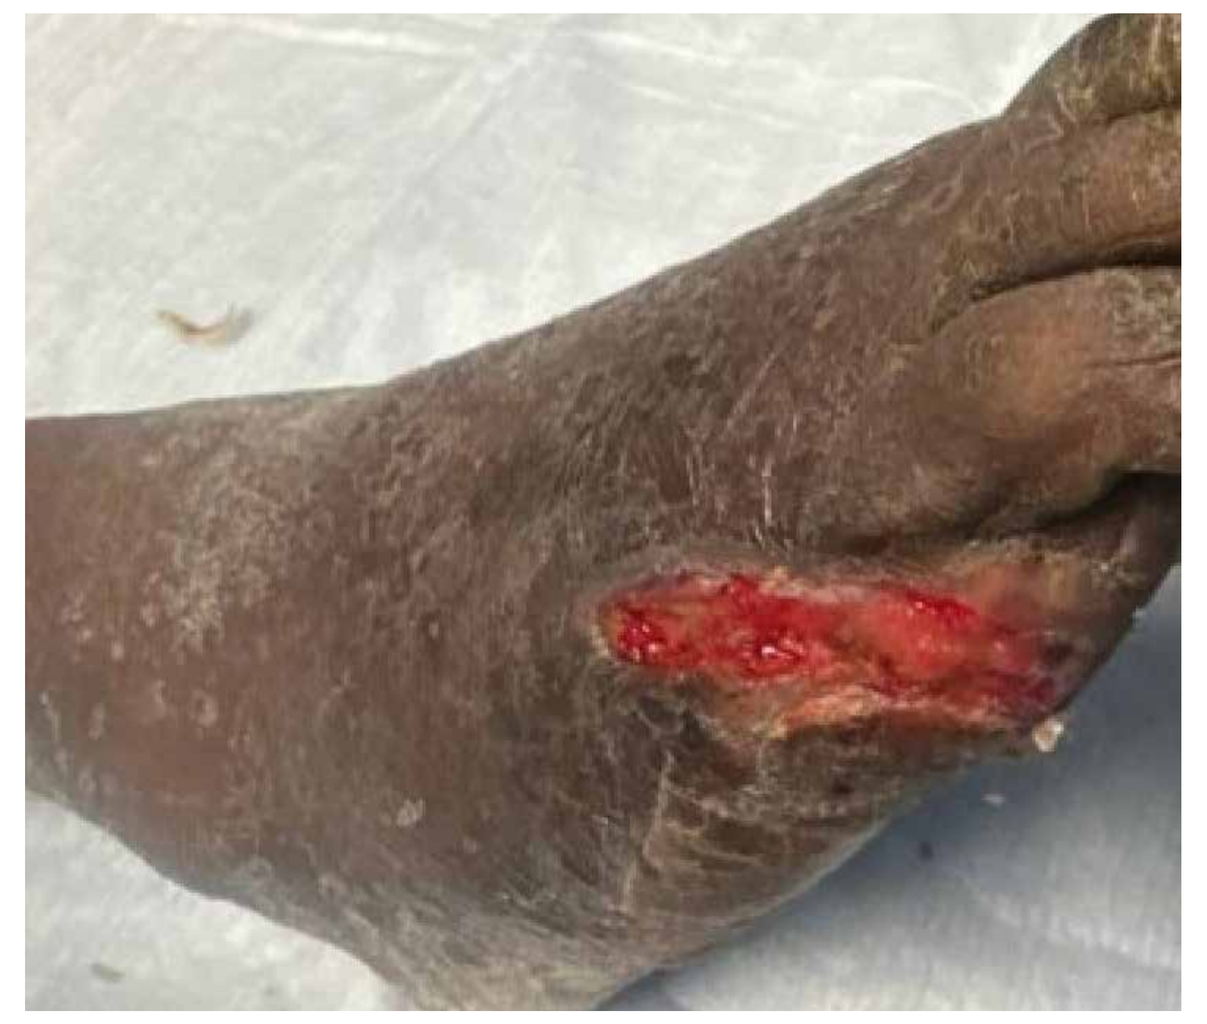

1/22/25 (4 WEEKS S/P DEBRIDEMENT AND APPLICATION OF KERECIS SKIN GRAFT

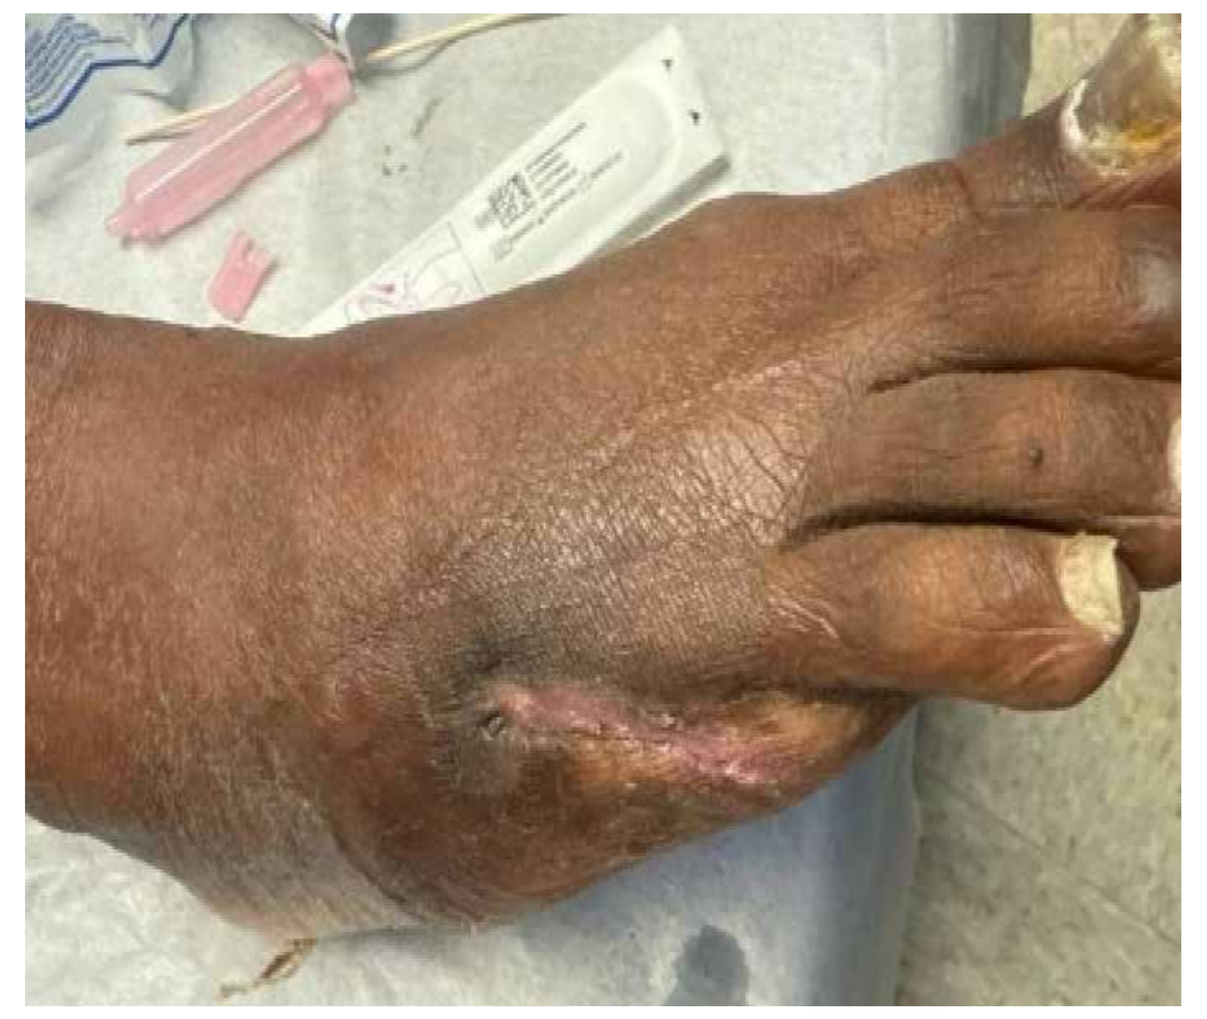

3/5/25 (10 WEEKS S/P DEBRIDEMENT AND APPLICATION OF KERECIS SKIN GRAFT)

RESULTS

All patients in the case series demonstrated progressive wound healing following the single application of Kerecis graft in combination with standard wound care and offloading. The wounds began to show evidence of granulation tissue formation and graft integration within the first 1–2 weeks post-application. No patients required a second graft or additional surgical intervention during the follow-up period.

By the third to fifth week, most wounds exhibited significant reduction in wound size and depth. Complete epithelialization and closure were achieved in all cases, with healing confirmed during weekly follow-up visits. The average time to full closure ranged from 4 to 6 weeks, depending on the initial wound size and location.

No adverse reactions to the Kerecis graft were observed. Additionally, there were no cases of wound infection, graft rejection, or further tissue breakdown during the course of treatment. All patients adhered to offloading instructions, which likely contributed to the positive outcomes.

Overall, the results suggest that a single application of Kerecis fish-skin graft, when combined with standard wound care and offloading, may be an effective and well-tolerated method to promote closure of deep diabetic foot wounds post-amputation and help prevent further complications.

CONCLUSIONS

Use of a single Kerecis fish-skin graft alongside standard wound care and consistent offloading supported successful healing of deep post-amputation diabetic foot wounds. Every patient in this series achieved full closure without complications, including infection, graft loss, or delayed healing. The graft's natural scaffold and omega-3–rich composition appeared to promote robust granulation and epithelial growth, contributing to accelerated wound improvement. Achieving timely closure helped protect against further tissue damage and lowered the likelihood of additional amputation. These findings indicate that Kerecis grafts may serve as an effective adjunct in limb-salvage strategies for high-risk diabetic individuals.

REFERENCES:

Kirsner, R. S., Marston, W. A., Snyder, R. J., Lee, T. D., & Cargill, D. I. (2020). Fish skin grafts in wound healing: A clinical update. Wound Repair and Regeneration, 28(5), 564–570. https://doi.org/10.1111/wrr.12814

Armstrong, D. G., Boulton, A. J. M., & Bus, S. A. (2017). Diabetic foot ulcers and their recurrence. New England Journal of Medicine, 376(24), 2367–2375. https://doi.org/10.1056/NEJMra1615439

Landsman, A., Molnar, J., & Shiloach, M. (2021). Clinical experience using fish skin graft for the treatment of chronic diabetic foot ulcers: A retrospective case series. Wounds, 33(4), 89–95.